Efficacy and Safety of Crisaborole in Patients With Mild-to-Moderate Atopic Dermatitis With and Without Food Allergies

Michael S. Blaiss,[1] Michael J. Cork,[2] Peter Lio,[3,4] Aharon Kessel,[5,6] Liza Takiya,[7] John L. Werth,[7] Michael A. O'Connell,[7] Chuanbo Zang,[7] Jonathan M. Spergel[8]

[1]Medical College of Georgia, Augusta University, Augusta, GA, USA; [2]Sheffield Dermatology Research, IICD, University of Sheffield, Sheffield Children's Hospital, Sheffield, United Kingdom; [3]Northwestern University Feinberg School of Medicine, Chicago, IL, USA; [4]Chicago Integrative Eczema Center, Chicago, IL, USA; [5]Bnai Zion Medical Center, Haifa, Israel; [6]Bruce and Ruth Rappaport Faculty of Medicine, Technion, Haifa, Israel; [7]Pfizer Inc., Collegeville, PA, USA; [8]Children's Hospital of Philadelphia and Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA

Copies of this poster obtained through this QR code are for your personal use only and may not be reproduced without permission from the authors.

Scan to download a reprint of this poster.

Copyright © 2021. All rights reserved.

Acknowledgments

Editorial/medical writing support under the guidance of the authors was provided by Christopher M. Goodwin, PhD, at ApotheCom, San Francisco, CA, USA, and was funded by Pfizer Inc., New York, NY, USA, in accordance with Good Publication Practice (GPP3) guidelines (*Ann Intern Med.* 2015;163:461-464).

Disclosures

MSB has served as an investigator or consultant for Pfizer and Regeneron/Sanofi Genzyme. MJC has served as an investigator or consultant for Astellas, Boots, Dermavant, Eli Lilly, Galapagos, Galderma, Hyphens, Johnson & Johnson, Kymab, L'Oreal, LEO, Menlo Therapeutics, Novartis, Oxagen, Perrigo (ACO Nordic), Pfizer, Procter & Gamble, Reckitt Benckiser, Regeneron/Sanofi Genzyme, and UCB Pharma. PL is a board member of the National Eczema Association; has served as an advisor or consultant for Regeneron/Sanofi Genzyme, Pfizer, Galderma, LEO, AbbVie, Eli Lilly, La Roche Posay, Pierre Fabre, Level Ex, Unilever, Menlo Therapeutics, Theraplex, IntraDerm, Exeltis, AOBiome, Arbonne, Amyris, Bodewell, Burt's Bees, and Kimberly Clark; holds stock in Altus Labs, Micreos, and Yobee Care; has received honoraria from UCB, Dermavant, Kiniksa, Verrica, Realm, and Johnson & Johnson, as well as fees from Regeneron/Sanofi Genzyme and Pfizer; and has served on speaker bureaus for Regeneron/Sanofi Genzyme, Pfizer, and Galderma. AK has served as an investigator or consultant for Pfizer, Sanofi, Takeda, Novartis, GSK, Rafa, AstraZeneca, and Kamada and has received honoraria or fees from Pfizer, Sanofi, Takeda, Novartis, GSK, Rafa, Teva, and Kamada. JMS has served as an investigator or consultant for Regeneron/Sanofi Genzyme, Celgene, Allakos, Novartis, and DBV Technology; has served on speaker bureaus for Abbot, Medscape, and Sanofi; and is an associate editor for the *Annals of Allergy, Asthma & Immunology*. LT, JLW, MAO, and CZ are employees and stockholders of Pfizer Inc.

Presented at the 2021 Winter Clinical Dermatology Conference; January 16-24, 2021

INTRODUCTION

- AD is a chronic inflammatory skin disease; patients with AD are at higher risk for other atopic comorbidities, such as food allergies[1]
- The prevalence of food allergy in AD ranges from 20% to 80% in this population[2]
 - Although it is not clear whether the presence of food allergies makes AD more difficult to treat, there is an association between ingestion of food that triggers an allergic reaction and AD exacerbation[3]
- Crisaborole ointment, 2%, is an anti-inflammatory nonsteroidal PDE4 inhibitor for the treatment of patients aged ≥3 months (≥2 years of age outside the United States) with mild-to-moderate AD[4]
 - Initial regulatory approval was based on the results from 2 identically designed, vehicle-controlled, phase 3 clinical studies: CORE 1 (NCT02118766) and CORE 2 (NCT02118792)[5]

OBJECTIVE

- **To ascertain the efficacy and safety of crisaborole for the treatment of AD in patients with or without food allergies in a post hoc pooled analysis from the phase 3 studies CORE 1 and CORE 2**

METHODS

Patients and Treatment

- CORE 1 and CORE 2 were 2 identically designed, randomized, double-blind, vehicle-controlled, phase 3 studies conducted to compare crisaborole and vehicle in patients with AD, per Hanifin and Rajka criteria,[6] who had mild-to-moderate disease per the ISGA and %BSA of ≥5 (excluding the scalp)
- Patients were randomly assigned in a 2:1 ratio to receive crisaborole ointment, 2%, or vehicle applied twice daily to all areas affected by AD, except the scalp, for 28 days
- Patients were not permitted to use any topical agents on AD lesions or rescue medications during the study
- For this post hoc analysis, patients were stratified based on their medical history of food allergies

Outcomes and Assessments

- The ISGA, a 5-point physician-reported scale of AD severity,[5] was assessed at baseline and weekly thereafter
- Pruritus severity was assessed using the SPS, a validated patient- or caregiver-reported 4-point rating scale,[7] and reported twice daily (morning and evening) via electronic diary
- Efficacy outcomes were
 - Proportion of patients who achieved ISGA success (defined as an ISGA of clear [0] or almost clear [1] with a ≥2-grade improvement from baseline) at day 29
 - Proportion of patients who achieved ISGA clear (0) or almost clear (1) at day 29
 - Proportion of patients who experienced improvement in SPS score (defined as a weekly average SPS score ≤1 point with ≥1-point improvement from baseline) at week 4
- Safety outcomes that included TEAEs (all cause and treatment related), serious AEs, and AEs of special interest (eg, anaphylaxis) were collected throughout the study

RESULTS

Patients

- In the pooled study population, 1016 patients received crisaborole and 506 received vehicle
 - Among them, 251 reported a past medical history of food allergies and 1271 did not have a past medical history of food allergies
 - Baseline demographics were generally similar between treatment arms and between those who did and those who did not have food allergies
 - Regarding baseline disease characteristics, for those with a past medical history of food allergies, a relatively greater proportion (1) used systemic corticosteroids previously, (2) used antihistamines concurrently, (3) had moderate AD per ISGA at baseline, and (4) had greater %BSA involvement with AD lesions (**Table 1**)

Table 1. Baseline Demographics and Disease Characteristics in Patients With and Patients Without Food Allergies

Demographic or Characteristic	HIstory of Food Allergies		No History of Food Allergies	
	Vehicle n=99	Crisaborole n=152	Vehicle n=407	Crisaborole n=864
Age, y				
Mean (SD)	8.3 (5.9)	10.8 (10.1)	13.0 (12.5)	12.6 (12.5)
Median (min, max)	8.0 (2, 36)	8.0 (2, 57)	10.0 (2, 79)	9.0 (2, 79)
Female, % (n)	51.5 (51)	45.4 (69)	56.5 (230)	57.5 (497)
White, % (n)	62.6 (62)	61.2 (93)	60 (244)	60.7 (524)
%BSA				
Mean (SD)	23.4 (20.9)	23.2 (21.9)	16.9 (16.1)	17.5 (17.1)
Median (min, max)	16.0 (5, 90)	15.0 (5, 90)	10.0 (5, 90)	10.0 (5, 95)
ISGA, % (n)				
Mild (2)	21.2 (21)	29 (44)	42.3 (172)	40.4 (349)
Moderate (3)	78.8 (78)	71.1 (108)	57.7 (235)	59.6 (515)
SPS, % (n)				
None (0)	0	3.3 (5)	4.7 (19)	3.5 (30)
Mild (1)	20.2 (20)	16.5 (25)	24.3 (99)	23.6 (204)
Moderate (2)	33.3 (33)	34.9 (53)	32.9 (134)	32.2 (278)
Severe (3)	30.3 (30)	30.3 (46)	26 (106)	30.3 (262)
Prior use of systemic corticosteroids,[a] % (n)	44.4 (44)	44.7 (68)	31.7 (129)	28.1 (243)
Concurrent use of antihistamines, % (n)	49.5 (49)	48.7 (74)	20.9 (85)	19.8 (171)

[a]Within 90 days before starting study treatment.

Efficacy

- The proportion of patients who achieved ISGA success at day 29 was significantly greater in the crisaborole-treated group than in the group receiving vehicle, regardless of past medical history of food allergies (**Figure 1**)
- Similarly, in patients with or without food allergies, a significantly greater proportion of crisaborole-treated patients achieved ISGA clear or almost clear at day 29 than those given vehicle (**Figure 2**)
- At week 4, a numerically greater proportion of patients with or without history of food allergies in the crisaborole group than in the vehicle group achieved improvement in SPS score; however, it was only statistically significant in the patients without history of food allergies (**Figure 3**)

Figure 1. Proportion of Patients Who Achieved ISGA Success[a] at Day 29

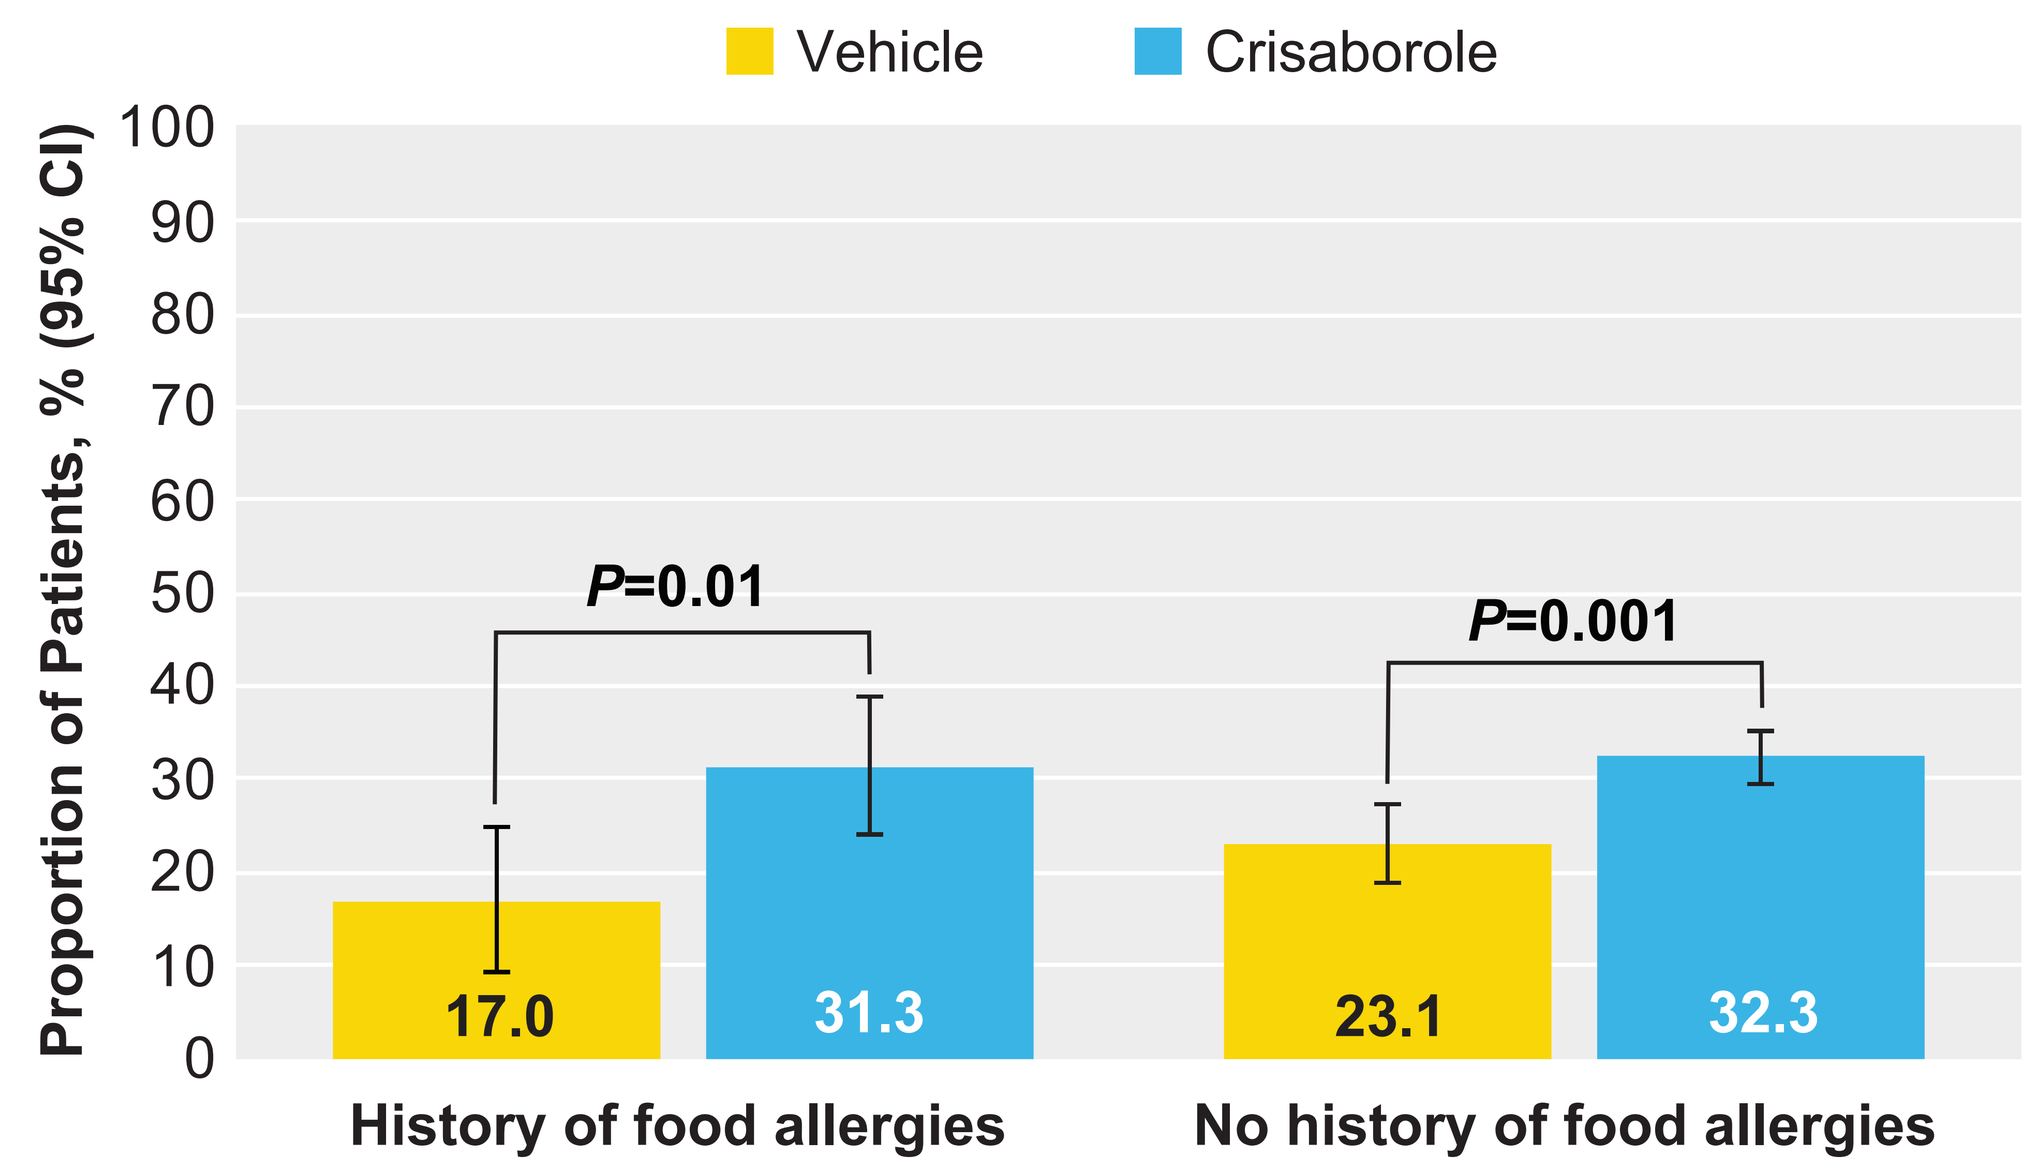
Vehicle
Crisaborole
Proportion of Patients, % (95% CI)
100
90
80
70
60
50
40
30
20
10
0
P=0.01
P=0.001
17.0
31.3
23.1
32.3
History of food allergies
No history of food allergies

[a]ISGA success defined as ISGA of clear or almost clear with ≥2-grade improvement from baseline.

Figure 2. Proportion of Patients Who Achieved ISGA Clear or Almost Clear at Day 29

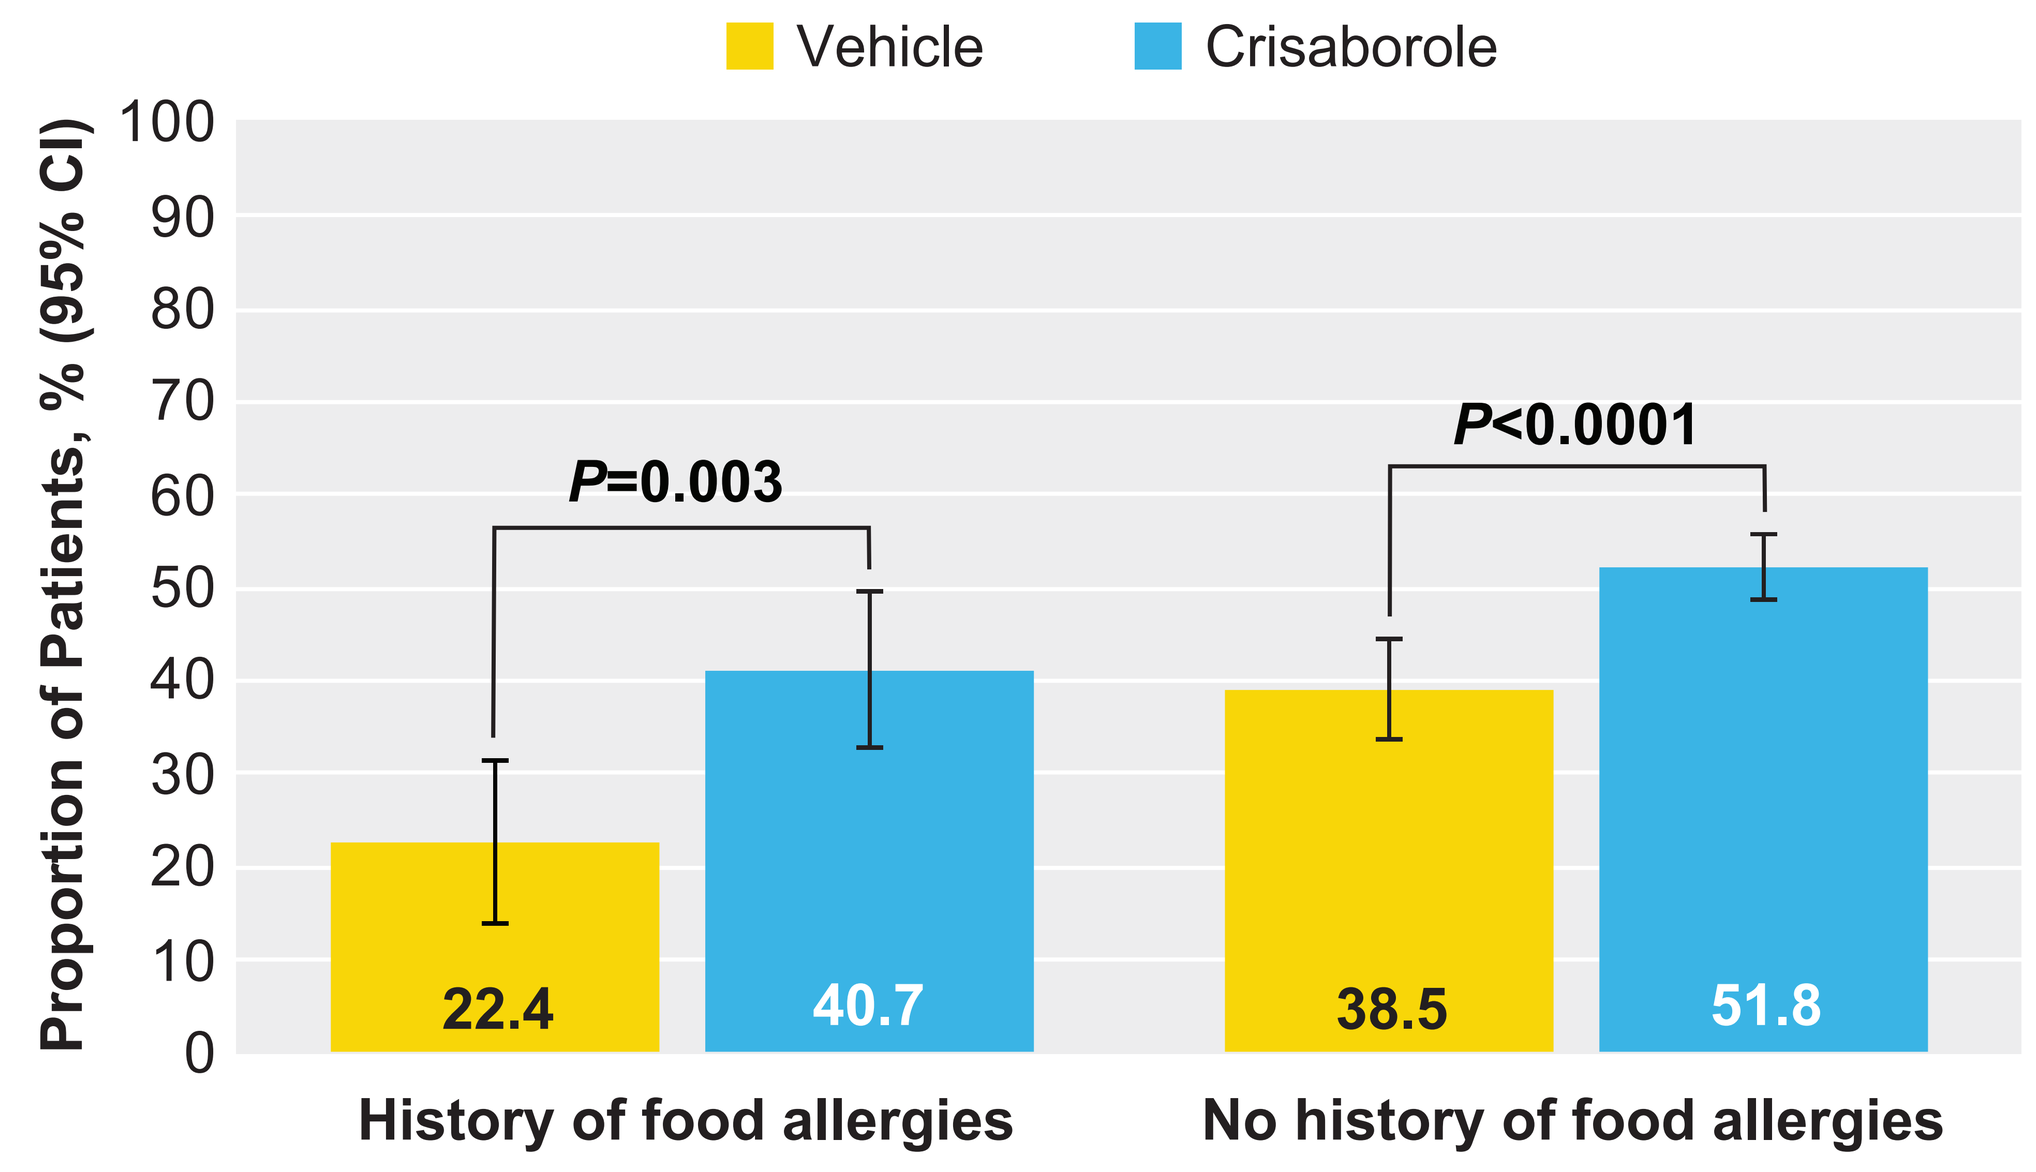
Vehicle
Crisaborole
Proportion of Patients, % (95% CI)
100
90
80
70
60
50
40
30
20
10
0
P=0.003
P<0.0001
22.4
40.7
38.5
51.8
History of food allergies
No history of food allergies

Figure 3. Proportion of Patients Who Achieved Improvement in SPS[a] Score at Week 4

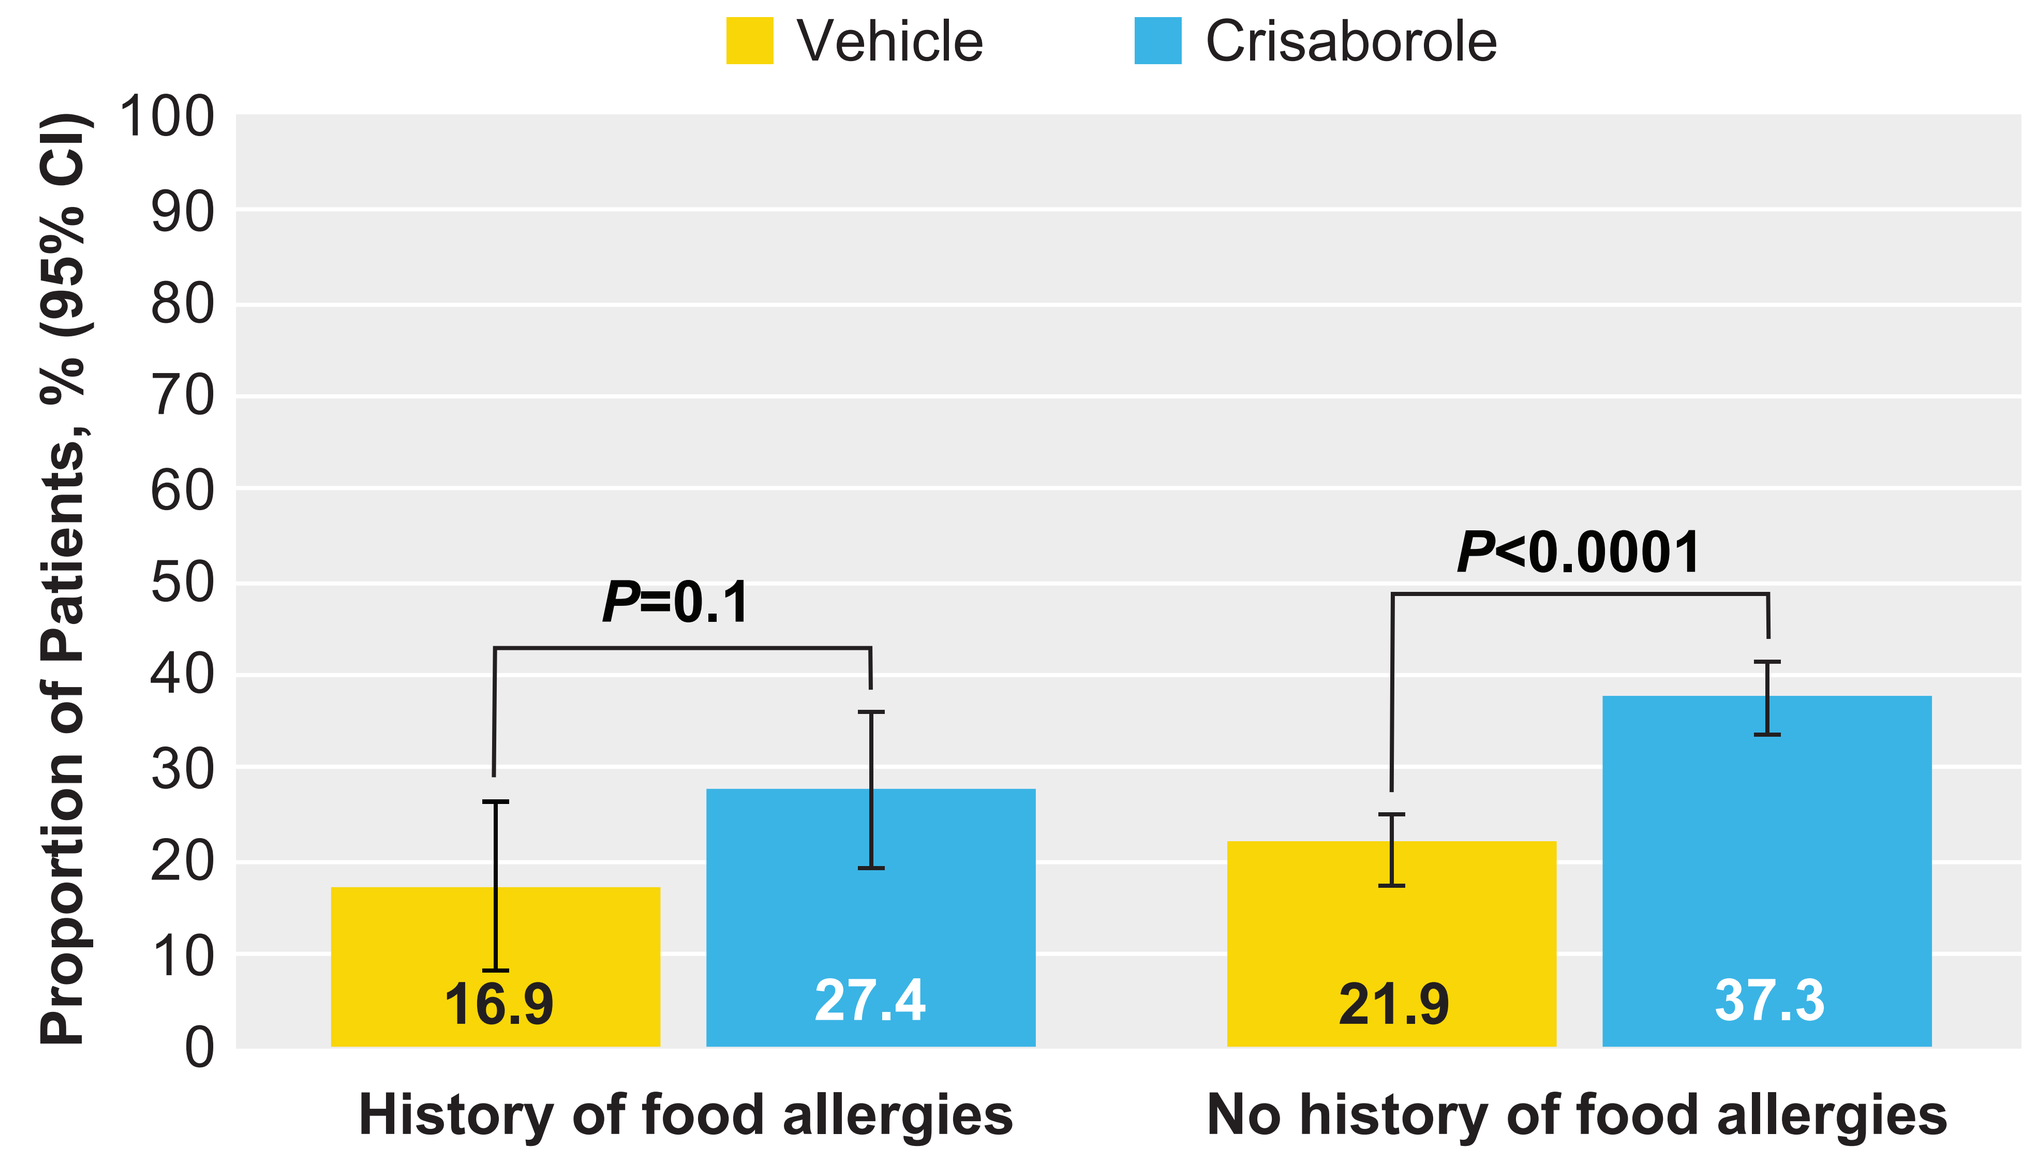
Vehicle
Crisaborole
Proportion of Patients, % (95% CI)
100
90
80
70
60
50
40
30
20
10
0
P=0.1
P<0.0001
16.9
27.4
21.9
37.3
History of food allergies
No history of food allergies

[a]Improvement in SPS score defined as weekly average SPS score ≤1 point with a ≥1-point improvement from baseline.

Safety

- The safety profile was generally similar between patients with food allergies and those without food allergies
- Among patients with food allergies, 61 crisaborole-treated patients (40.1%) and 40 vehicle-treated patients (40.8%) experienced at least 1 all-cause TEAE
 - Among crisaborole-treated patients with food allergies, 22 (14.5%) experienced a mild TEAE, 37 (24.3%) experienced a moderate TEAE, and 2 (1.3%) experienced a severe TEAE
- Individual TEAEs were infrequent (**Table 2**)
- The most common treatment-related TEAE in patients with food allergies and those without food allergies (crisaborole vs vehicle) was application site pain (7.2% vs 3.1% and 4.0% vs 0.7%, respectively)
- 1 crisaborole-treated patient with food allergies experienced a serious TEAE (pneumonia, 0.7%), and no patients (0%) given vehicle experienced a serious TEAE; the serious TEAE was not considered related to treatment
- No anaphylaxis was reported in any group

Table 2. Most Common (in >2% Patients) TEAEs (all cause) in Patients With and Patients Without Food Allergies

	History of Food Allergies		No History of Food Allergies	
	Vehicle n=98	Crisaborole n=152	Vehicle n=401	Crisaborole n=860
AE (all cause), % (n)				
Application site pain	3.1 (3)	7.2 (11)	0.7 (3)	4.0 (34)
Upper respiratory tract infection	1.0 (1)	5.3 (8)	4.0 (16)	3.0 (26)
Viral upper respiratory tract infection	1.0 (1)	2.0 (3)	2.5 (10)	2.7 (23)
Pyrexia	3.1 (3)	2.6 (4)	1.2 (5)	2.3 (20)
Eczema infected	1.0 (1)	2.0 (3)	0.2 (1)	0.1 (1)
Cellulitis	2.0 (2)	0.7 (1)	0.2 (1)	0.1 (1)
Staphylococcal skin infection	2.0 (2)	0.7 (1)	0.7 (3)	0
Asthma	1.0 (1)	2.6 (4)	0	0.5 (4)
Cough	5.1 (5)	1.3 (2)	1.0 (4)	1.6 (14)
Nasal congestion	2.0 (2)	3.3 (5)	0	0.5 (4)
TEAEs of special interest, % (n)				
Anaphylaxis	0	0	0	0

Limitations

- These post hoc analyses were not powered to detect treatment differences in food allergy subgroups; therefore, additional studies are necessary to confirm these results
- The CORE-1 and CORE-2 studies were not specifically designed to evaluate crisaborole effects on food allergies

CONCLUSIONS

- **Regardless of whether patients have food allergies, crisaborole is effective in treating mild-to-moderate symptoms of AD**
- **The safety profile was generally similar between patients with food allergies and those without food allergies; no new safety signals were observed**
- **Crisaborole should be considered for management of AD in patients with or without a history of food allergies**

Abbreviations **%BSA**, percentage of treatable body surface area; **AD**, atopic dermatitis; **AE**, adverse event; **ISGA**, Investigator's Static Global Assessment; **PDE4**, phosphodiesterase 4; **SPS**, Severity of Pruritus Score; **TEAE**, treatment-emergent adverse event.

References **1.** Dharmage SC et al. *Curr Opin Allergy Clin Immunol.* 2004;4(5):379-385. **2.** Werfel T, Breuer K. *Curr Opin Allergy Clin Immunol.* 2004;4(5):379-385. **3.** Robison RG, Singh AM. *J Allergy Clin Immunol Pract.* 2019;7(1):35-39. **4.** Eucrisa (crisaborole) ointment, 2%, for topical use [package insert]. New York, NY: Pfizer Labs; April 2020. **5.** Paller AS et al. *J Am Acad Dermatol.* 2016;75(3):494-503.e496. **6.** Hanifin JM, Rajka G. *Acta Derm Venereol.* 1980;60(92):44-47. **7.** Yosipovitch G et al. *Itch.* 2018;3:e13.

This study was funded by Pfizer Inc.